Real-World robustness of B/F/TAF at virological failure in the ANRS-CO3-AquiVIH-NA cohort

739

Pantxika Bellecave[1], Alaric Peyrouny-Mazeau[2], Olivier Leleux[2], M. Hessamfar[2,3], G. Le Moal[4], D. Neau[5], L. Alleman[6], C. Cazanave[5], E. Lazaro[7], P. Duffau[3], M-A Vandenhende[3], Bernard Castan[8], Fabrice Bonnet[3], **Camille Tumiotto[1,9]**

[1]CHU Bordeaux, Service de Virologie, Hôpital Pellegrin, Bordeaux, France, [2]Bordeaux Population Health Research Center, INSERM U1219, CIC-EC 1401, Univ. Bordeaux - ISPED, Bordeaux, France; [3]CHU de Bordeaux, Service de Médecine Interne et Maladies Infectieuses, Hôpital Saint-André, Bordeaux, France; [4]CHU de Poitiers, Service de Maladies Infectieuses et Tropicales, Poitiers, France; [5]CHU de Bordeaux, Service de Maladies Infectieuses et Tropicales, Hôpital Pellegrin, Bordeaux, France; [6]CH de la Côte Basque, Service de Maladies Infectieuses, Bayonne, France; [7]CHU de Bordeaux, Service de Médecine Interne et Maladies Infectieuses, Hôpital Haut-Lévêque, Pessac, France; [8]CH de Périgueux, Service de Maladies Infectieuses et Tropicales, Périgueux, France
[9]CNRS-UMR 5234 Microbiologie Fondamentale et Pathogénicité, Université de Bordeaux, Bordeaux, France

BACKGROUND

- Clinical trials demonstrated the robustness and the high efficacy of Bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF).
- Real-world evidence data in people living with HIV-1 (PLH) experiencing virologic failure (VF) on B/F/TAF remains limited, especially on emerging resistance associated mutations (RAMs).
- **Objective: To retrospectively describe the virological outcomes in pre-treated PLH who experienced VF on B/F/TAF, based on their genotypic data and their viral load at treatment initiation**

METHODS

- HIV-1 adults from the regional prospective cohort ANRS-CO3-AquiVIH-NA (France) treated with B/F/TAF between 2018/01/01 and 2021/12/31 were included if they had:
 - a HIV-1 viral load (VL) available within 1 year
 - a CD4 measurement available within 2 years
 - at least one genotypic resistance test (GRT) available before B/F/TAF initiation.
- Virological data:
 - VF was defined as either 2 consecutive HIV-1 viral loads (VL)>50 copies/mL or 1 VL>200 copies/mL
 - GRTs were performed using Sanger sequencing as part of standard clinical care. RAMs were reported according to the v34 ANRS resistance algorithm (https://hivfrenchresistance.org/)).
 - Virological outcomes at the last follow-up visit: B/F/TAF discontinuation/ continuation, last VL

RESULTS

Study population

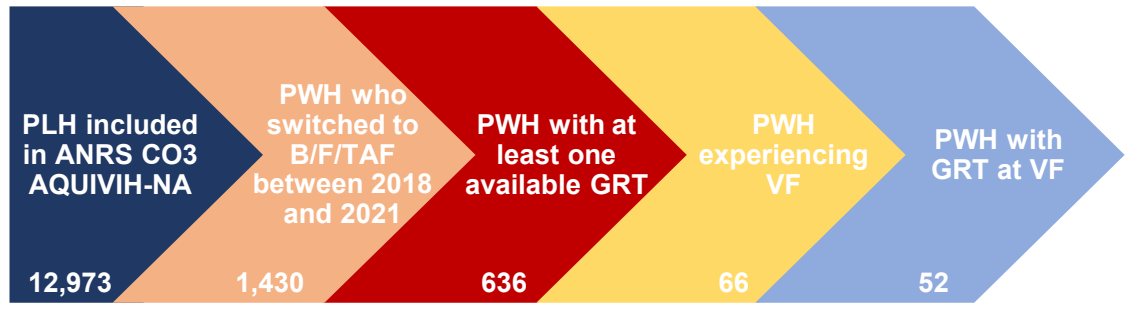
PLH included in ANRS CO3 AQUIVIH-NA
12,973
PWH who switched to B/F/TAF between 2018 and 2021
1,430
PWH with at least one available GRT
636
PWH experiencing VF
66
PWH with GRT at VF
52

**Emergence of resistance occurred for 6/66 (9.1%) pre-treated PLH with documented virological failure on B/F/TAF.
Viral suppression was observed on 71.4% PLH following B/F/TAF despite previous VF, suggesting that the main cause of VF on this regimen could be due to poor adherence issues.**

Characteristics of PLH experiencing Virological Failure

	PWH with baseline VL<50 n= 31	PWH with baseline VL>50 N= 35	PWH total N=66
Age, years, median [min-max]	48.8 [23.8-68.6]	54,6 [30.8-74.4]	54.4 [23.8-74.4]
Previous cART, number of lines	3 [1-12]	7 [1-13]	4 [1-13]
Previous VF, n (%)	21 (67.7)	35 (100)	56 (84.8)
Baseline VL, copies/mL, median [min-max]	28 [0- 49]	140 [53-599,503]	57 [0- 599,503]
Baseline CD4, cells/µL, median [min-max]	669 [93-1721]	622 [41-1630]	638 [41-1721]
Baseline B/F/TAF RAMs providing resistance to: n (%):			
3TC/FTC	8 (25.8)	16 (45.7)	24 (36.4)
TDF/TAF	3 (9.7)	5 (14.3)	8 (12.1)
BIC	0 (0.0)	0 (0.0)	0 (0.0)
VF VL[1], copies/mL, median [min-max]	640 [52-567,564]	213 [57-340,000]	258 [52-567,564]
Delay before VF[2], months [min-max]	16 [2-59]	13 [5-54]	13 [2-59]

[1] 2nd VL > 50 copies/mL measurement or 1st VL measurement >200 copies/mL. [2] delay between B/F/TAF initiation and 2nd VL > 50 copies/mL or 1st VL >200 copies/mL

Emerging RAMs of PLH with baseline VL>50 copies/mL

#	Clade	Baseline RAMs: VL (copies/mL)	Baseline RAMs: previous NRTI RAMs	Baseline RAMs: Previous INSTIs RAMs	Baseline RAMs: GSS	VF RAMs: Delay to VF (months)	VF RAMs: VL (copies/mL)	VF RAMs: VF NRTI RAMs	VF RAMs: VF INSTIs RAMs	VF RAMs: GSS*
1	B	95	0	*No data*	3	13	297	0	0	3
2	B	8530	184I	0	2	10	304	0	0	3
3	B	140	184V	*No data*	2	35	578	184V	0	2
4	B	599,503	184V	0	2	5	617	*No data*	*No data*	*ND*
5	B	322	41L, 74V, 184V, 215FL	*No data*	1.5	46	4,520	70KT, 184VM, 215LF	0	2
6	B	238	0	0	3	6	181	GRT failed	0	3
7	B	81	41L, 210W, 215Y	0	2.5	6	263	0	0	3
8	B	72	69NS, 70R, 184V	0	2	12	57	69NTS, 70RK, 184VM	0	2
9	B	260	0	0	3	11	156	0	0	3
10	B	127	184V	0	2	43	126	*No data*	*No data*	*ND*
11	B	7,310	41L, 184V, 210W, 215Y	0	1.5	11	48,400	0	0	3
12	B	58	41L, 184V, 210W, 215Y	0	1.5	16	74	*No data*	*No data*	*ND*
13	**B**	**371**	**0**	**0**	**3**	**54**	**77**	**184V**	GRT failed	**2**
14	01_AE	123,000	0	*No data*	3	22	22,800	0	0	3
15	B	192	184V	0	2	13	69	0	0	3
16	B	125,000	69N, 184IM	0	2	45	100,000	0	0	3
17	B	55	0	0	3	15	340,000	0	0	3
18	B	75	210W 215D	0	3	6	91	210W 215D	GRT failed	3
19	B	58	0	*No data*	3	35	69	0	0	3
20	G	67	184V	0	2	19	232	0	0	3
21	URF	778	0	0	3	7	312	0	0	3
22	B	86	0	0	3	12	121	0	0	3
23	B	53	41L, 67N, 184V, 210W, 215Y	0	1	5	190	GRT failed	0	3
24	**URF**	**273,000**	**67N 184V T215I K219Q**	**0**	**2**	**11**	**213**	**184V**	**263K**	**1**
25	B	257	0	0	3	19	190	*No data*	*No data*	*ND*
26	B	80	67N 184V 210W 215Y	0	1.5	12	139	0	GRT failed	3
27	F1	94	0	0	3	17	88	0	0	3
28	02_AG	356	184V	0	2	8	95	184VM	0	2
29	B	68,900	41W, 184V, 215TA	0	2	17	18,200	0	0	3
30	F1	12,400	0	0	3	14	146,172	0	0	3
31	B	87	0	0	3	11	89	0	GRT failed	3
32	D	109	44D	0	3	8	128	GRT failed	0	3
33	02_AG	146	210M	0	3	15	247	210M	0	3
34	A2	90	69N	0	3	22	271	0	0	3
35	B	79	41LM, 44KE, 67DG, 215DA	0	3	7	219	GRT failed	0	3

ND: not determined; GSS: Genotypic Sensitivity Score (Sensitivity = 1; partially resistant = 0.5; resistant =0). *GSS at VF was calculated according to VF GRT

Emerging RAMs of PLH with baseline VL<50 copies/mL

#	Clade	Baseline RAMs: Previous NRTI RAMs	Baseline RAMs: Previous INSTIs RAMs	Baseline RAMs: GSS	VF RAMs: VF VL (copies/mL)	VF RAMs: Delay to VF (months)	VF RAMs: VF NRTI RAMs	VF RAMs: VF INSTIs RAMs	VF RAMs: GSS*
36	B	41L, 184IM, 215EK	0	2	20,940	12	*No data*	*No data*	*ND*
37	02_AG	0	*No data*	3	35,065	3	*No data*	*No data*	*ND*
38	B	69TA	0	3	4,410	8	GRT failed	0	3
39	02_AG	67G, 70T, 184V, 215F	74ML 97A	2	32,400	13	GRT failed	0	3
40	B	0	0	3	52	30	*No data*	*No data*	*ND*
41	B	0	0	3	567,564	20	*No data*	*No data*	*ND*
42	A1	0	0	3	9,350	36	GRT failed	0	3
43	B	0	0	3	145	12	*No data*	*No data*	*ND*
44	B	41L, 184V, 210W, 215Y	*No data*	1.5	289	25	*No data*	*No data*	*ND*
45	B	0	0	3	239	6	*No data*	*No data*	*ND*
46	13_cpx	69TI	74I, 145PS	3	253	34	*No data*	*No data*	*ND*
47	URF	0	0	3	761	3	*No data*	*No data*	*ND*
48	URF	0	0	3	446	9	*No data*	*No data*	*ND*
49	URF	69+, 70R, 215N	0	1	199,000	54	69+, 70R, 215N	0	1
50	**02_AG**	**0**	**0**	**3**	**222,000**	**28**	**184VM**	GRT failed	2
51	B	215NY, 219KN	0	3	89	18	0	0	3
52	**B**	**0**	**0**	**3**	**78**	**4**	**184V**	**0**	**2**
53	**B**	**0**	**0**	**3**	**750**	**43**	**184V**	**155NH**	**1**
54	URF	184I	0	2	4,070	18	0	0	3
55	G	0	0	3	2,230	11	0	0	3
56	A3	0	0	3	1,500	42	0	0	3
57	B	0	0	3	210	2	0	0	3
58	**C**	**0**	**0**	**3**	**640**	**9**	**184V**	**0**	**2**
59	B	0	0	3	210	29	0	0	3
60	B	0	0	3	118	22	0	0	3
61	02_AG	67ND, 69TS, 70RK, 184VM, 219KQ	0	2	110	16	67ND, 69TS, 70R, 184VM, 219Q	0	2
62	B	0	0	3	106	4	0	0	3
63	B	41L, 67N, 70R, 184V, 210W, 215E, 219Q	0	1	293	59	41L, 67N, 70R, 184V, 210W, 215E, 219Q	0	1
64	B	74V, 184V	0	2	926	6	0	0	3
65	B	0	0	3	238	38	0	0	3
66	A1	0	0	3	8,560	11	0	0	3

ND: not determined; GSS: Genotypic Sensitivity Score (Sensitivity = 1; partially resistant = 0.5; resistant =0). *GSS at VF was calculated according to VF GRT

Follow-up of PLH after Virological failure

- B/F/TAF was maintained for 56 participants (including #13, #50 and #52 with emerging M184V) and 40 of them (71,4%) had a HIV-1 VL <50 copies/mL at their last visit.
- ARV treatment was modified to PI- or NNRTI-based regimens for 6 participants (# 23, 24, 26, 34, 53 and 58); Doravirine was added to B/F/TAF for 1 (#35)
- No follow-up was available for 3 participants: #17 was lost for follow-up and #29 and #30 died

CONCLUSIONS

- Among PLH with documented VF on B/F/TAF, emergence of resistance occurred in 6/66 (9.1%). Selected RAMs were five M184V, one N155H and one R263K. Preexisting NRTIs RAMs did not seem to enable additional RAMs at failure.
- Poor or inconsistent adherence may explain most of VF cases, as 71,4% of participants pursuing B/F/TAF after VF achieved viral suppression. This emphasizes the importance of adherence counselling in improving outcomes.

ACKNOWLEDGMENTS

Scientific committe : P. Bellecave, P. Blanco, F. Bonnet (Chair), S. Bouchet, D. Breilh, C. Cazanave, S. Desjardin, V. Gaborieau, A. Gimbert, M. Hessamfar, L. Lacaze-Buzy, ME. Lafon, E. Lazaro, O. Leleux., G. Le Moal, D. Malvy, P. Mercié, D. Neau, I. Pellegrin, A. Perrier, V. Petrov-Sanchez, A. Peyrouny-Mazeau, C. Tumiotto, M.O. Vareil, L. Wittkop (Methodologist); *Participating centers :* Hôpital Saint André, CHU de Bordeaux, Médecine Interne et Maladies Infectieuses, (N. Bernard, F. Bonnet, D. Bronnimann H. Chaussade, D. Dondia, P. Duffau, I. Faure, V. Hémar, M. Hessamfar, W. Jourde, C. Martell, P Mercié, A. Monier, P. Morlat, E. Mériglier, F. Paccalin, R. Paz, E. Ribeiro, C. Rivoisy, MA Vandenhende); Hôpital Pellegrin, CHU de Bordeaux, Maladies Infectieuses et Tropicales, (L. Barthod, C. Cazanave, FA. Dauchy, A. Desclaux, H. Dutronc, A. Duvignaud, J. Leitao, D. Neau, D. Nguyen, D. Malvy, T. Pistone, M. Puges, C. Tinevez, C. Vignal, G. Wirth); Hôpital Haut-Lévêque, CHU de Bordeaux, Médecine Interne et Maladies Infectieuses, (C. Courtault, F. Camou, C. Greib, E. Lazaro, JL. Pellegrin, C. Prot-Leurent, E. Rivière, JF. Viallard); Hôpital d'Agen, Médecine Interne (E. Klément-Fructos, A. Nonis, P. Rispal); Hôpital de Libourne, Médecine Interne (O. Caubet, S. De Faucal, M. Favarel-garrigues, H. Ferrand, S. Tchamgoué); Hôpital de Bayonne, Maladies Infectieuses (M. Acquier, L. Alleman, S. Farbos, MO. Vareil, H. Wille); Hôpital de Dax, Médecine Interne et Maladies Infectieuses, (K. André, L. Caunegre, Y. Gérard, M. Lauda-Maillen); Hôpital Saint-Cyr/Villeneuve-sur-Lot, Maladies Infectieuses, (I. Chossat); Hôpital de Mont de Marsan, Médecine Interne et Maladies Infectieuses, (Y. Gerard, F. Lacassin, C. Lasbasses-Depis); Hôpital d'Arcachon, Médecine Interne, (A. Barret, M. Bouet, C. Courtault, M. Roucoules, M. Videcoq); Hôpital de Périgueux, Médecine Interne et Maladies Infectieuses, (C. Belzunce, B. Castan, J. Koffi, N. Rouanes, A. Saunier, J.B. Zabbe); Hôpital de Pau, Médecine Interne et Maladies Infectieuses, (G. Dumondin, V. Gaborieau); Hôpital d'Orthez, Médecine Interne, (Y. Gerard) ; CHU de Poitiers, Médecine Interne et Maladies Infectieuses, (G. Beraud, G. Le Moal, M. Catroux, M. Garcia, V. Giraud, JP. Martellosio, F. Roblot, JM. Turmel) ; Hôpital de Saintes, Médecine Interne, (T. Pasdeloup) ; Hôpital d'Angoulême, Médecine Interne, (A. Riché, M. Grosset, S. Males, C. Ngo Bell) ; Hôpital de Jonzac, Maladies Infectieuses, (T. Pasdeloup), Hôpital de Saint Jean d'Angely, Maladies Infectieuses, (T. Pasdeloup) ; Hôpital de Royan, Médecine Interne, (P. Mottaz, T. Pasdeloup) ; Hôpital de Châtellerault, Médecine Interne (A, Elsendoorn); *Other departements :* Immunology: P. Blanco, I. Pellegrin ; CRB-BBS: C. Carpentier, I. Pellegrin ; Virology: P. Bellecave, C. Tumiotto ; Pharmacology: S. Bouchet, D. Breilh, G. Miremont-Salamé ; Data collection : G. Arnou, A. Barhoumi, P. Camps, S. Delveaux, S. Lawson-Ayayi, E. Lenaud, D. Plainchamps, A. Pougetoux, I. Stragier, B. Uwamaliya, K. Zara; IT department: V. Conte, M. Gapillout ; Project Team : O. Leleux (Project Leader), A. Perrier (Data Manager), A. Peyrouny-Mazeau (Statistitien) ; Participants of the Cohort ANRS CO3-AQUIVIH-NA.

Sponsor: CHU de Bordeaux
FUNDING: ANRS| MIE, CHU de Bordeaux and GILEAD Sciences Inc.