Digital Strategy to improve PrEP adherence among MSM: A Stepped-wedge Randomized Trial in China

1131

Chunyan Li[1,2], Zhuoheng Yin[1], Quanmin Li[3], Linghua Li[3], Songjie Wu[4], Ke Liang[4], Gifty Marley[1], Chengxin Fan[1], Yifan Dai[1], Haojie Huang[5], Aniruddha Hazra[6], Jonathan Lio[6], Joseph D. Tucker[7,8], Renslow Sherer[6], **Weiming Tang[1,7]**

[1]UNC Project China, Guangzhou, China, [2]Tokyo College, The University of Tokyo, Tokyo, Japan, [3]Guangzhou Eighth People's Hospital, Guangzhou, China, [4]Zhongnan Hospital of Wuhan University, Wuhan, China, [5]Wuhan Tongxing LGBT Center, Wuhan, China, [6]University of Chicago, Chicago, IL, USA [7]University of North Carolina at Chapel Hill, Chapel Hill, NC, USA, [8]London School of Hygiene and Tropical Medicine, London, UK,

BACKGROUND

- Digital Health intervention is promising for encouraging behavior change.
- We conducted a stepped-wedge trial (SWT) to evaluate the efficacy of co-created digital PrEP adherence intervention to facilitate adherence among Chinese MSM PrEP users.

METHODS

- The SWT is nested in a PrEP demonstration trial in Guangzhou and Wuhan, China (NCT04754139).
- MSM in Wuhan and Guangzhou were recruited by online ads, clinic flyers, and community referrals to a PrEP demonstration trial. MSM were prescribed TDF/TFC as oral PrEP and could opt for daily or on-demand regimens.
- Behavioral survey data were collected at baseline and quarterly follow-ups over 12 months (M3, M6, M9, M12).
- Per-protocol analysis and generalized linear mixed-effects model were conducted in Stata 15.0.

Intervention development:

1. **Before the trial:** Open call to solicit ideas of digital PrEP adherence messages
2. **During the trial:** 19 trial participants joined the co-creation group

 Co-creation: Online and off-line sessions to iteratively develop digital messages for PrEP adherence
3. The co-created messages were sent to participants every other week through WeChat (a mobile chat app).

- Intervention messages were co-created through open calls and three co-creation rounds, including short videos, images, and infographics.

RESULTS（July 2021 to September 2024）

- 1,134 MSM started oral PrEP, with a median age of 29.1(SD=5.9).
- 1,089 MSM enrolled the SWT trial. A total of 7 people were diagnosed of HIV.
- 388 (34.2%) lost to follow-up before the trial ended; no difference across SWT clusters.
- About half of the participants opted for on-demand PrEP (the “2+1+1” strategy).
- From M3-M12, about 75% reported optimal adherence (daily users: 6-7 pills/week; on-demand users: full compliance with the 2-1-1 dosing scheme per sex event within 30 days; people not having sex considered as adherent), with a non-significant secular trend of declining adherence over time.
- The digital intervention did not significantly improve adherence levels (aOR=1.20, 95% C.I.= 0.88-1.63)

Table 1. Percentages of participants reporting optimal PrEP adherence across clusters over time

SWT cluster (baseline N)	M3 (n=906)	M6 (n=827)	M9 (n=759)	M12 (n=710)
Cluster A (n=247)	77.7%	80.7%	72.9%	75.2%
Cluster B (n=284)	77.1%	81.0%	77.3%	73.9%
Cluster C (n=278)	77.7%	73.6%	77.5%	78.0%
Cluster D (n=280)	80.8%	78.5%	74.9%	75.8%
ALL (n=1089)	78.4%	78.4%	75.8%	75.8%

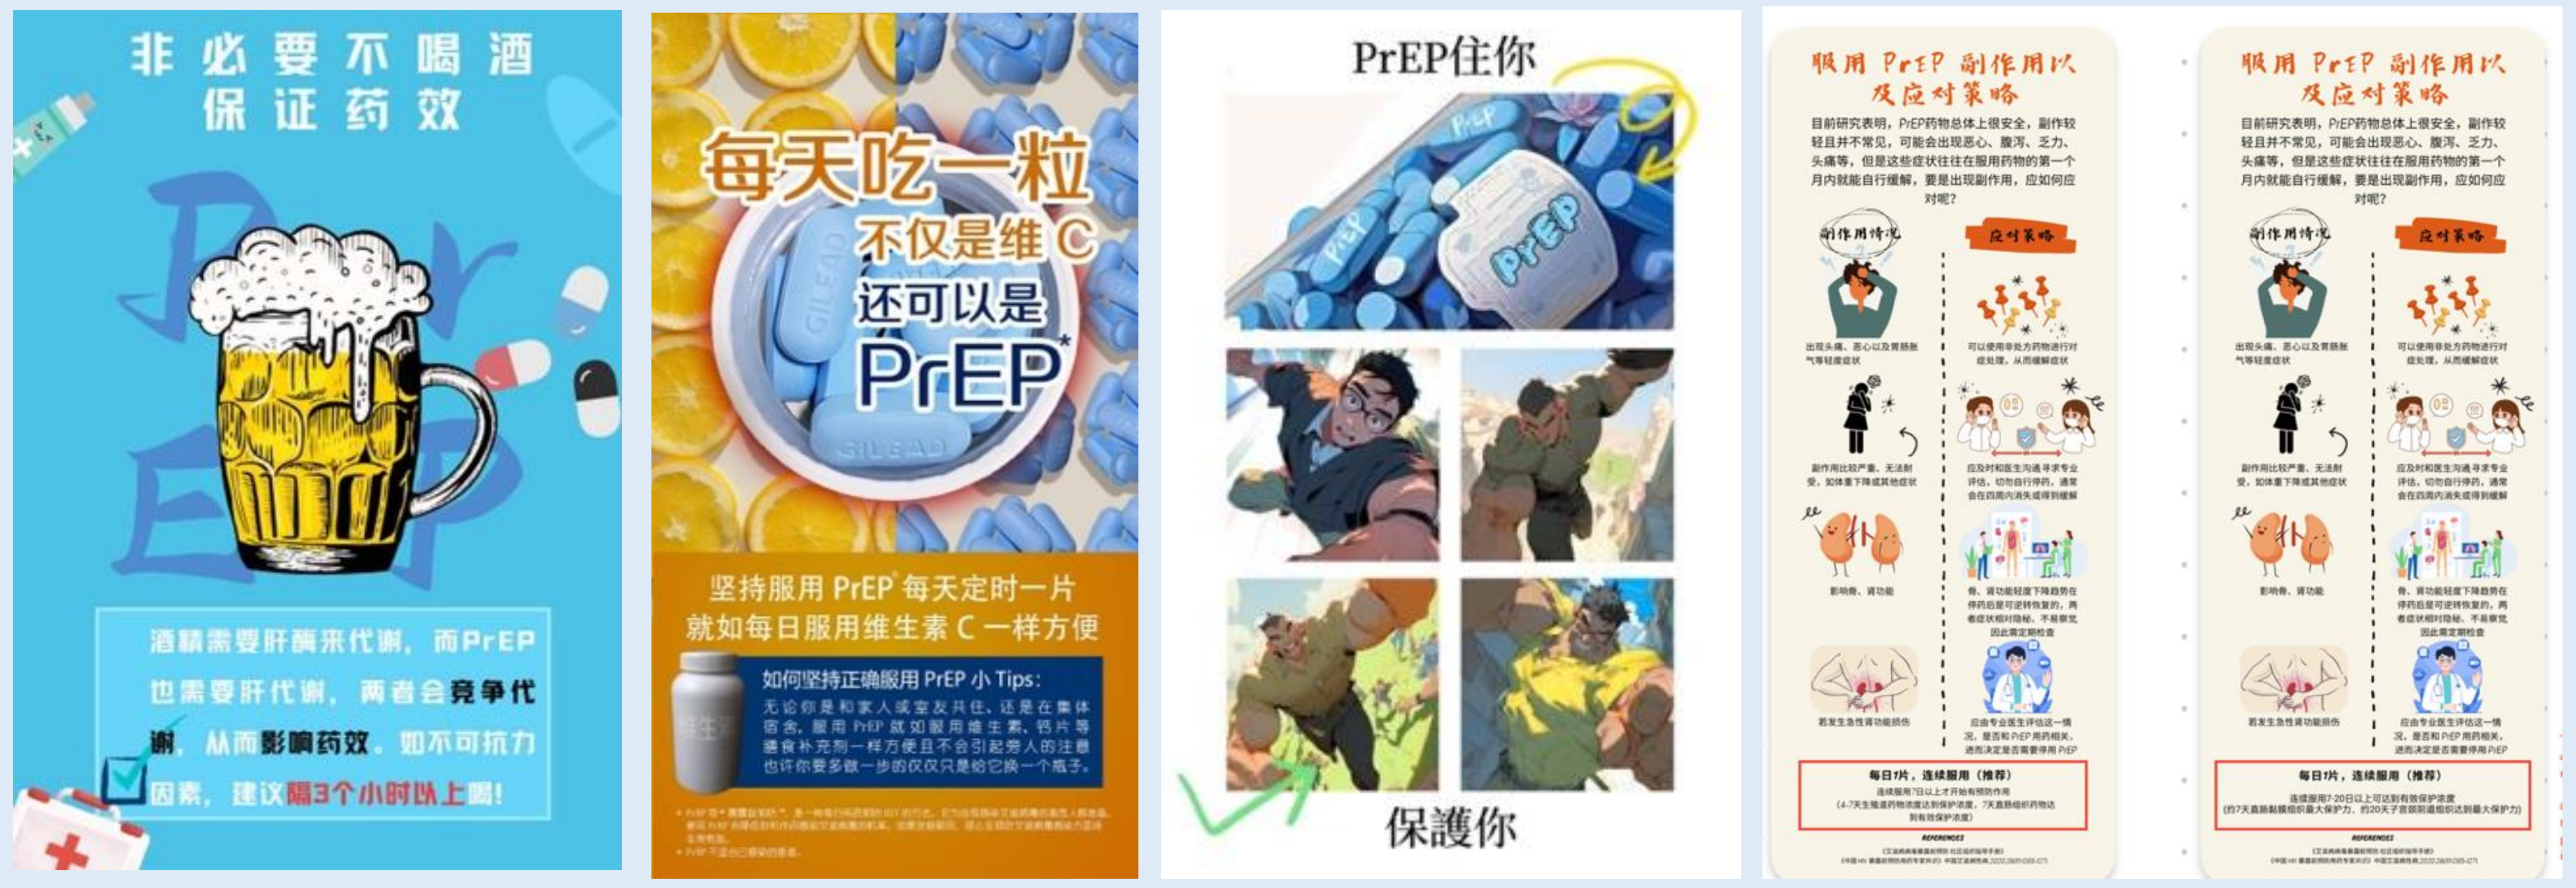
非必要不喝酒
保证药效
酒精需要肝酶来代谢，而PrEP也需要肝代谢，两者会竞争代谢，从而影响药效。如不可抗力因素，建议隔3个小时以上喝！
每天吃一粒
不仅是维C
还可以是
PrEP
坚持服用 PrEP 每天定时一片
就如每日服用维生素 C 一样方便
如何坚持正确服用 PrEP 小 Tips:
PrEP住你
保護你
服用PrEP副作用以及应对策略
服用PrEP副作用以及应对策略

Figure 1. Adjusted Odds Ratio for predictors of optimal PrEP adherence – results from GLMM

	Adjusted incidence ratio (95% CI)
Intervention (ref: no)	1.01 (0.91-1.13)
App use frequency (ref: no use)	
> Once a week	1.10 (0.95-1.27)
> Once a month	1.05 (0.96-1.15)
Perceived app help (ref: no help)	
Somewhat helpful	0.95 (0.84-1.08)
Very helpful	1.06 (0.94-1.21)
Event-driven dosing (ref: once daily)	0.65 (0.60-0.71)*

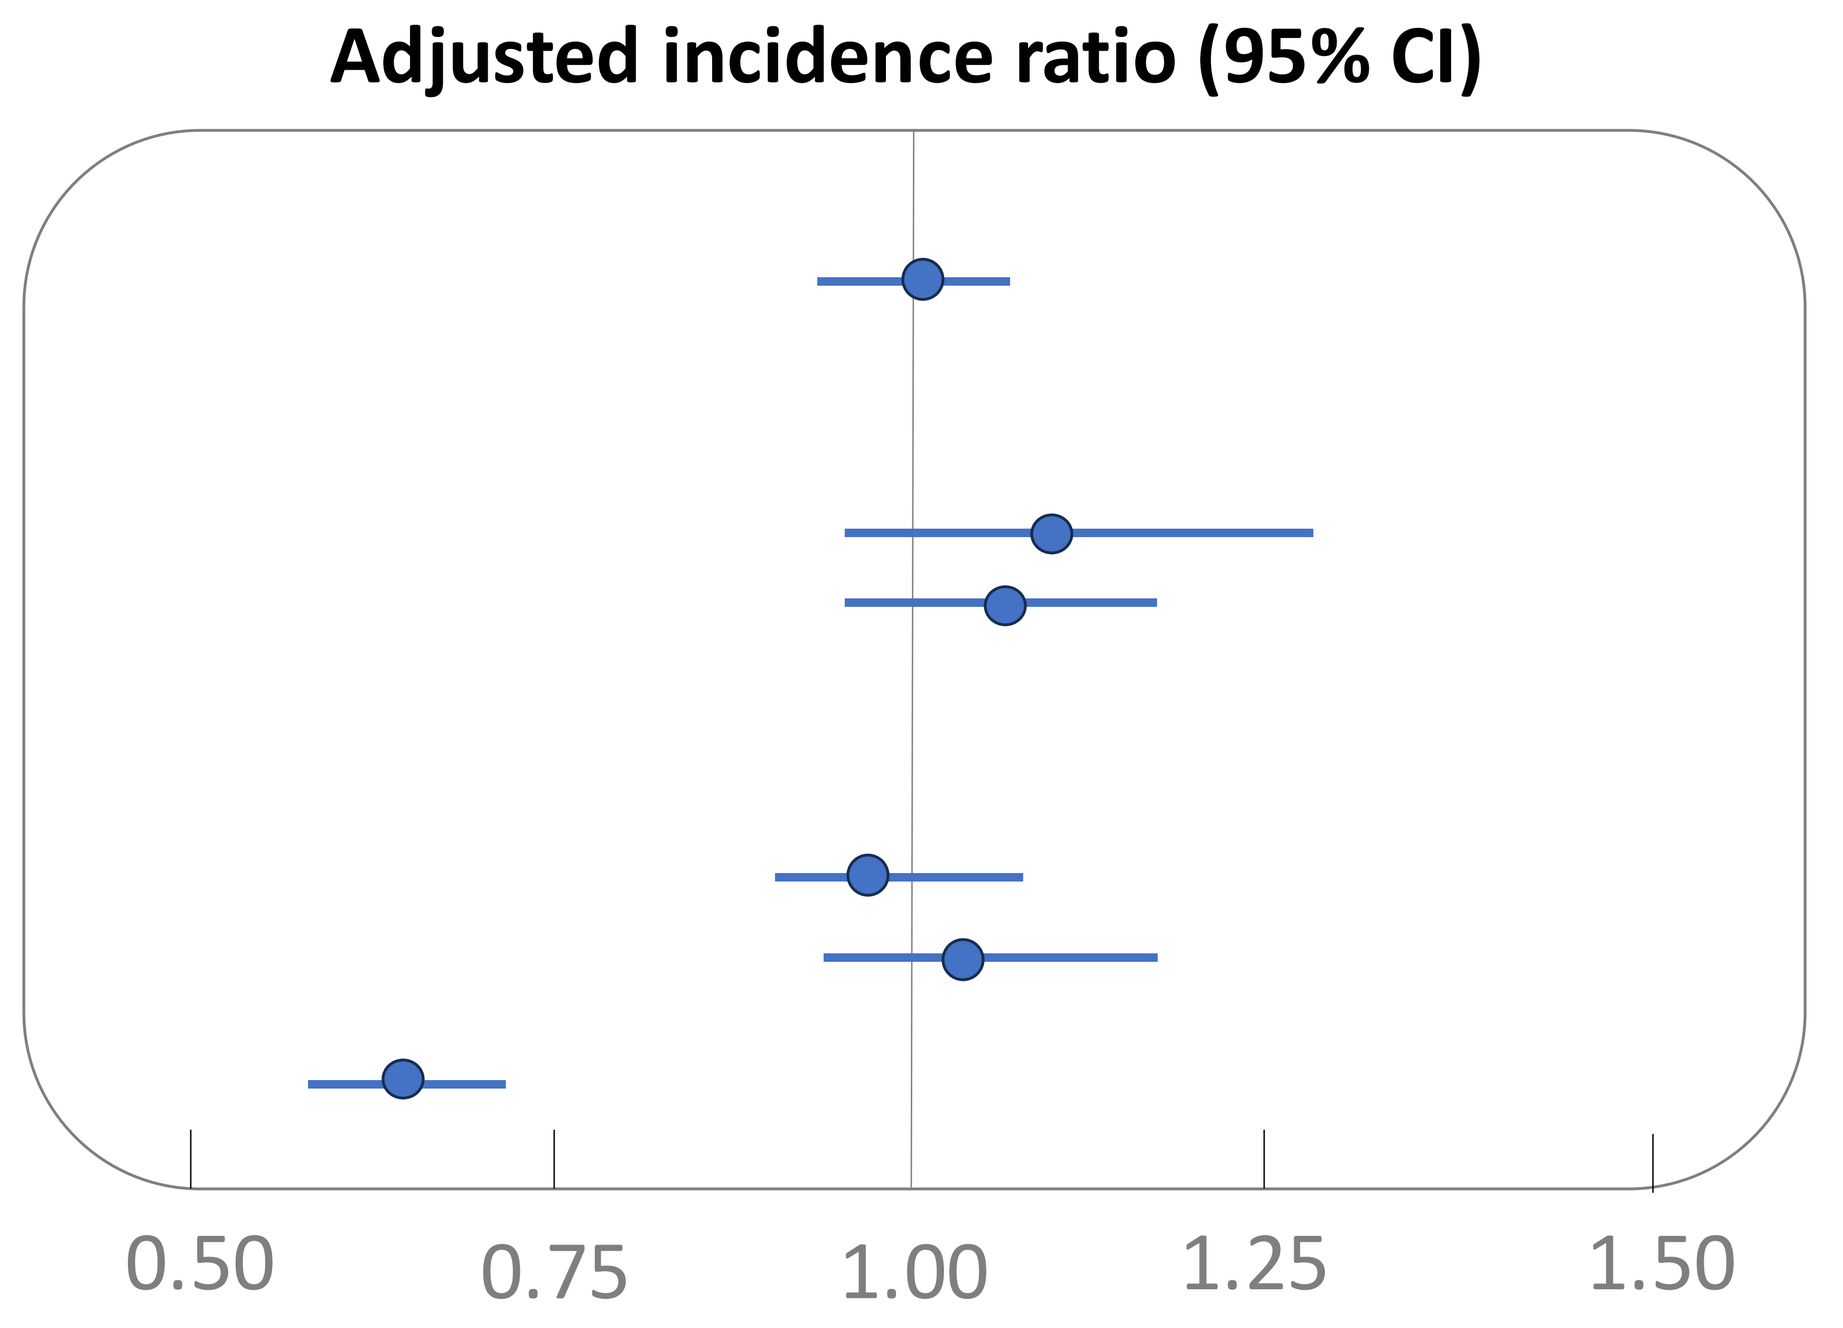
Adjusted incidence ratio (95% CI)
0.50
0.75
1.00
1.25
1.50

CONCLUSIONS

- MSM using once-daily oral regimen are more likely to report optimal medication adherence.
- Exposure to the digital co-created message, or higher frequency of using the intervention app, or higher perceived help from the intervention app did not significantly improve medication adherence.
- Co-created digital PrEP adherence intervention with Chinese MSM has the potential to improve PrEP adherence.
- Message-based intervention may have limited impact on improving adherence behaviors.
- Further research is needed to analyze the relationship between behavior outcomes and the extent of actual intervention exposure (i.e. reviewing the message).
- Our findings of varied adherence performance among individuals highlight the need for future research into differentiated PrEP interventions.

For further information, please contact

Chunyan Li (chunyanli@alumni.unc.edu)
Weiming Tang (weiming_tang@med.unc.edu)
Renslow Sherer, Jr. (rsherer@uchicago.edu)

Acknowledgements:

We would like to thank all other investigators from Guangzhou Eighth People's Hospital (Heping Zhao, Cong Liu, Xu Yang, Dandan Gong); Social Entrepreneur to Spur Health (Hengran Qian, Shixiang Bao, Yingge Wang, Nachuan Wu, Jiayi Luo, Bin Yu, Yutong Jiang, Yongjie Sha, Yuan Xiong, Anna Fan), Kun Wang (Wuhan Tongzhi Center), Jie Liu (Zhongnan Hospital of Wuhan University), Ivy Jiang (the University of Chicago).

This research was funded by Gilead Sciences Inc.

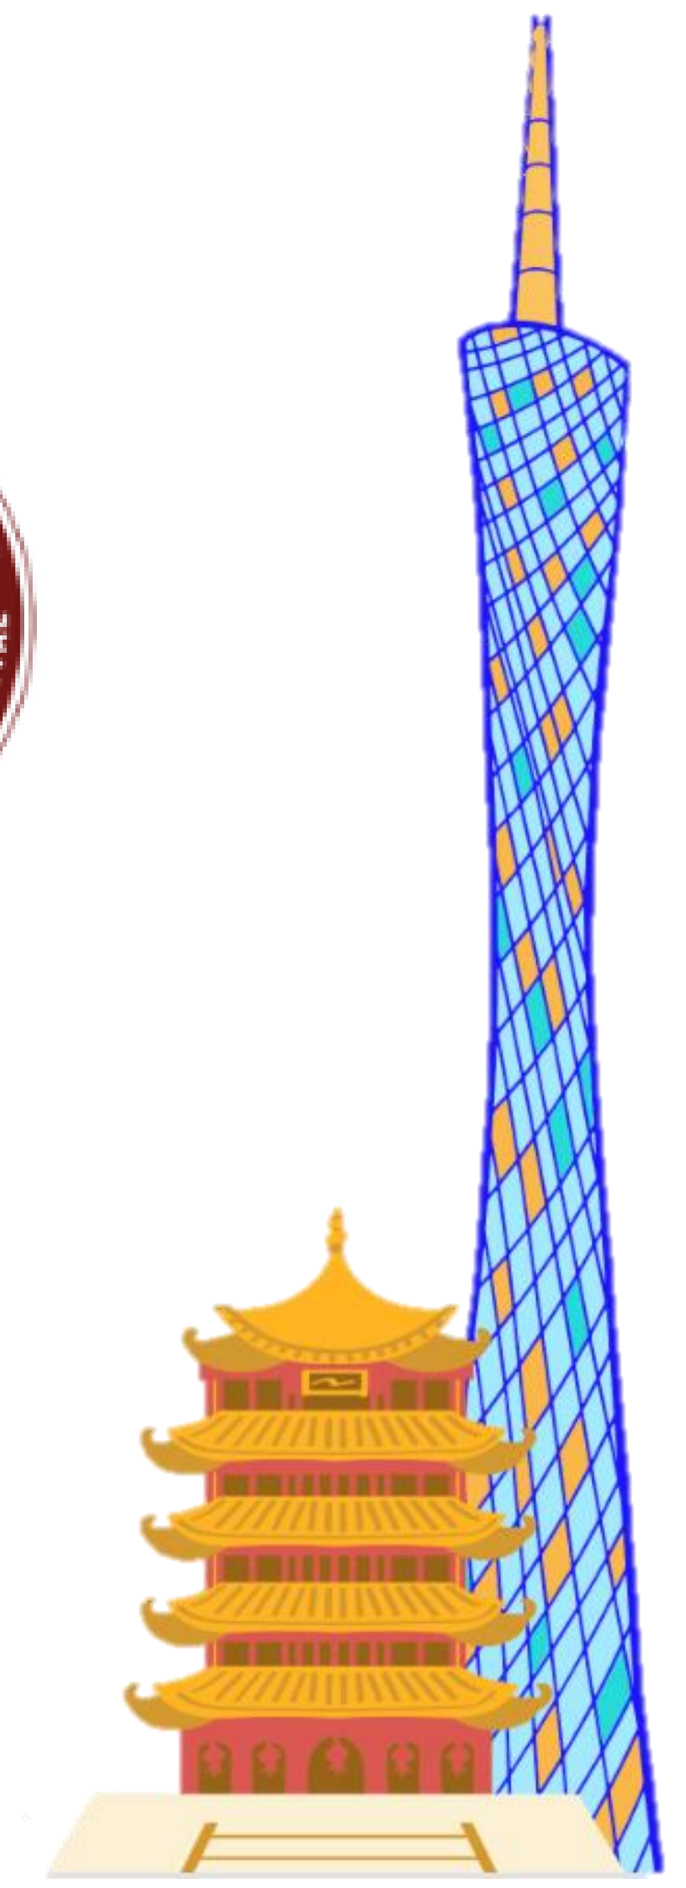